STIMULATED TUMOR CELLS (STC) VACCINE INDUCE RESPONSE IN COLORECTAL CANCER

#1132

G. ALZEEB[1], C. GONGORA[2], C. TORTORELLI[1], C. RICHARD[3], R. BOIDOT[3], T. FORTIN[4], A. BESSEDE[5], Y. WANG[6], L. CHALUS[1], B. PINTEUR[1], P. BRAVETTI[1], A. ITALIANO[7]

1.Brenus Pharma, Lyon, France; 2. Institut de Recherche en Cancérologie de Montpellier, Montpellier, France; 3.UMR CNRS 6302, Dijon, France; 4.Anaquant, Lyon, France; 5.Explicyte, Bordeaux, France; 6. Inovotion, La Tronche, France; 7. Institut Bergonié, Bordeaux, France.

STC-1010 : A HUMAN THERAPEUTIC VACCINE TO TREAT PATIENTS WITH COLORECTAL CANCER

COLORECTAL CANCER (CRC) : 2nd CAUSE OF CANCER MORTALITY

2 Millions new cases diagnosed / year - 3rd most common cancer type

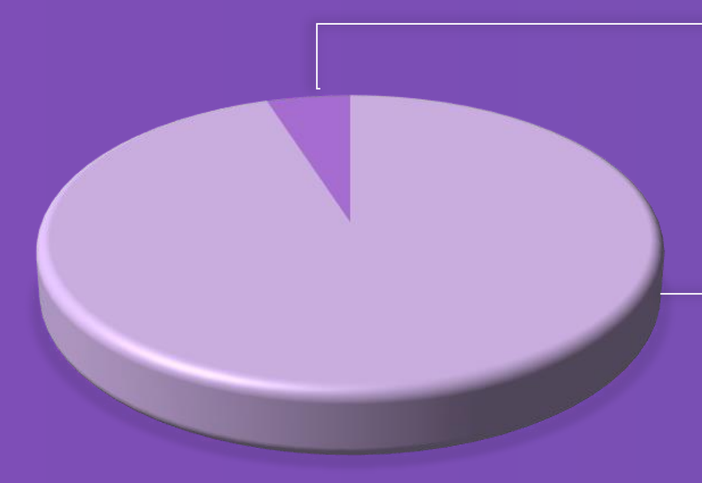

5% of mCRC patients : MSI-H Population

95% of mCRC patients : MSS Population
Treated by chemotherapy in 1L (5FU, OX, IR)

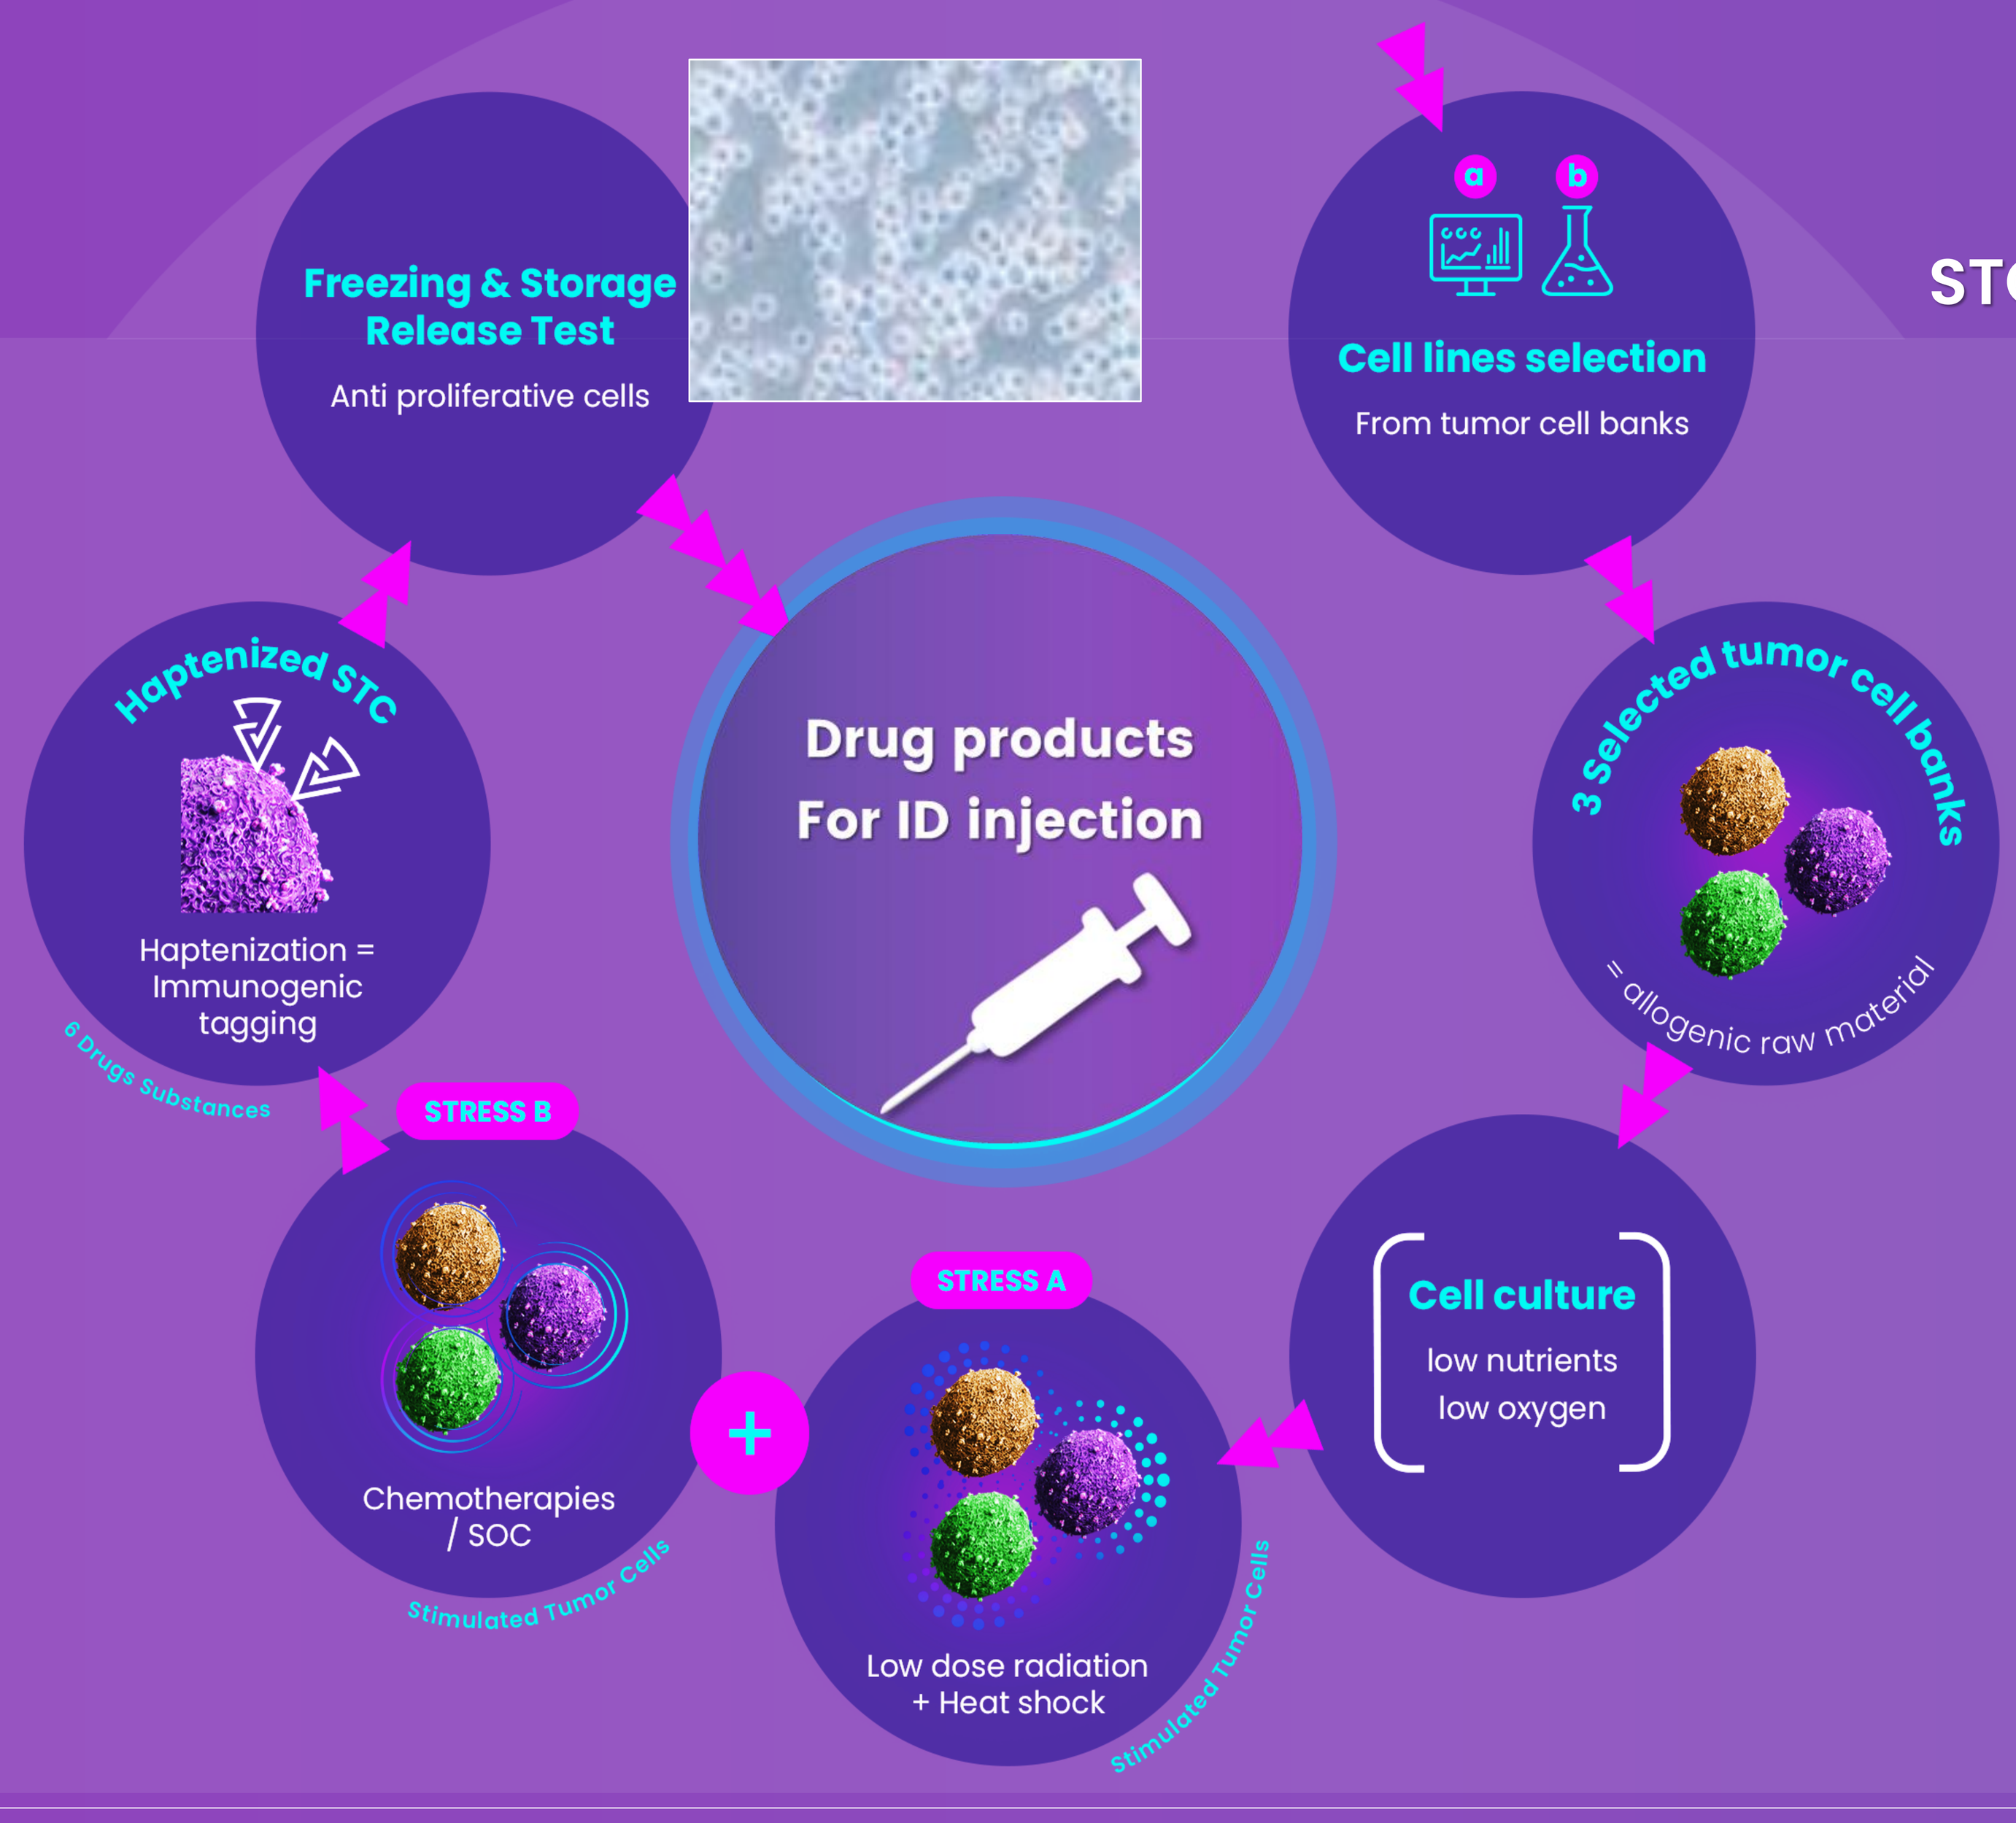
Freezing & Storage Release Test
Anti proliferative cells
Cell lines selection
From tumor cell banks
3 Selected tumor cell banks
= allogenic raw material
Cell culture
low nutrients
low oxygen
STRESS A
Low dose radiation + Heat shock
Stimulated Tumor Cells
STRESS B
Chemotherapies / SOC
Stimulated Tumor Cells
Haptenized STC
Haptenization = Immunogenic tagging
6 Drugs Substances
Drug products
For ID injection

STC-1010 BIOPRODICTION PROCESS

1. DATA-MINING SELECTION
a. Database screening b. In vitro testing

Selection criteria to cover most of CRC phenotypes and clinical heterogeneity:

- Representative oncogenic drivers
- Cold & Hot tumor cell lines
- Resistance capability

STC drug product is made up of 3 CRC cell lines well characterized

- **HCT116** (carcinoma), **HT29** (adenocarcinoma, **LoVo** (adenocarcinoma)

2. STRESS EXPOSITION

The 3 cell lines are stressed under metabolic conditions to mimic anti-angiogenic treatment with nutrients depletion

- **Physical stress** (A) :
 25 (Gy) and heat shock
- **Chemical stress** (B)
 CRC Standards chemotherapy :
 Oxaliplatin, 5-FU, Irinotecan

Reproduce antigenic tumoral signature linked to treatment resistances

3. HAPTENIZATION PROCESS with DNFB*

Covalent linkage to safely induce immunogenicity of the Stimulated Tumor Cells

STC-1010
« GHOST » TUMOR CELLS STRESSED & HAPTENIZED
3 DS (A)+ 3 DS(B) = 6 DS* = DP*

DNFB : dinitrofluorobenzene, DS : Drug Substances, DP : Drug Product

VACCINE CHARACTERIZATION

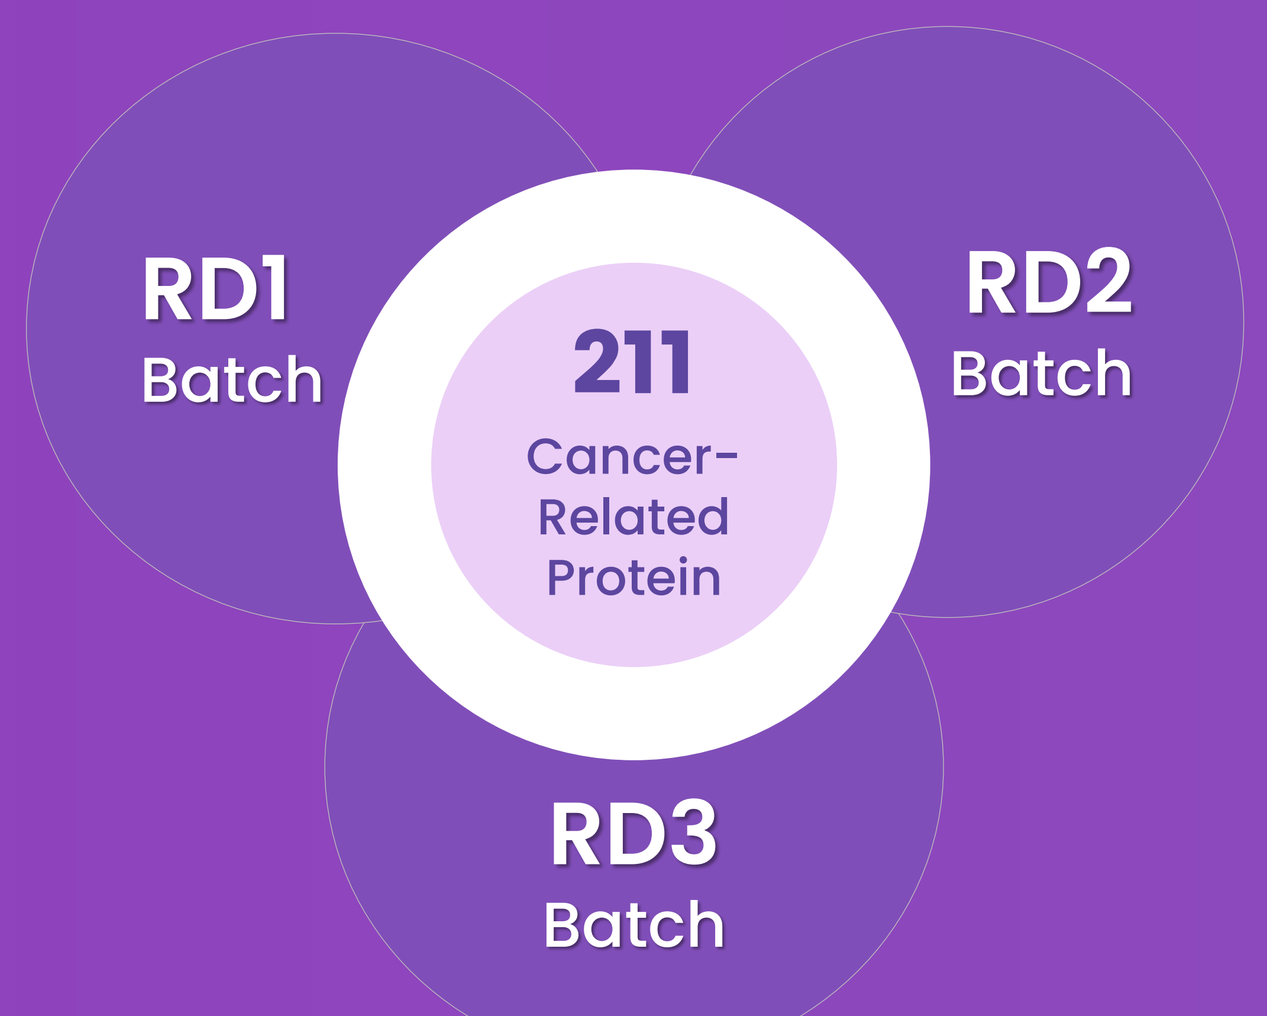
RD1 Batch
RD2 Batch
211 Cancer-Related Protein
RD3 Batch

Fig 1. LC/MS-MS Proteomic analysis of STC batches

RD BATCH 3	
Drug Substance	Specific protein identified by LC/MS Analysis
DSA HCT116	46
DSB HCT116	50
DSA HT29	80
DSB HT29	56
DSA LOVO	24
DSB LOVO	99

RD1	Tox batch
RD2	Optimisation batch
RD3	Scale-up batch

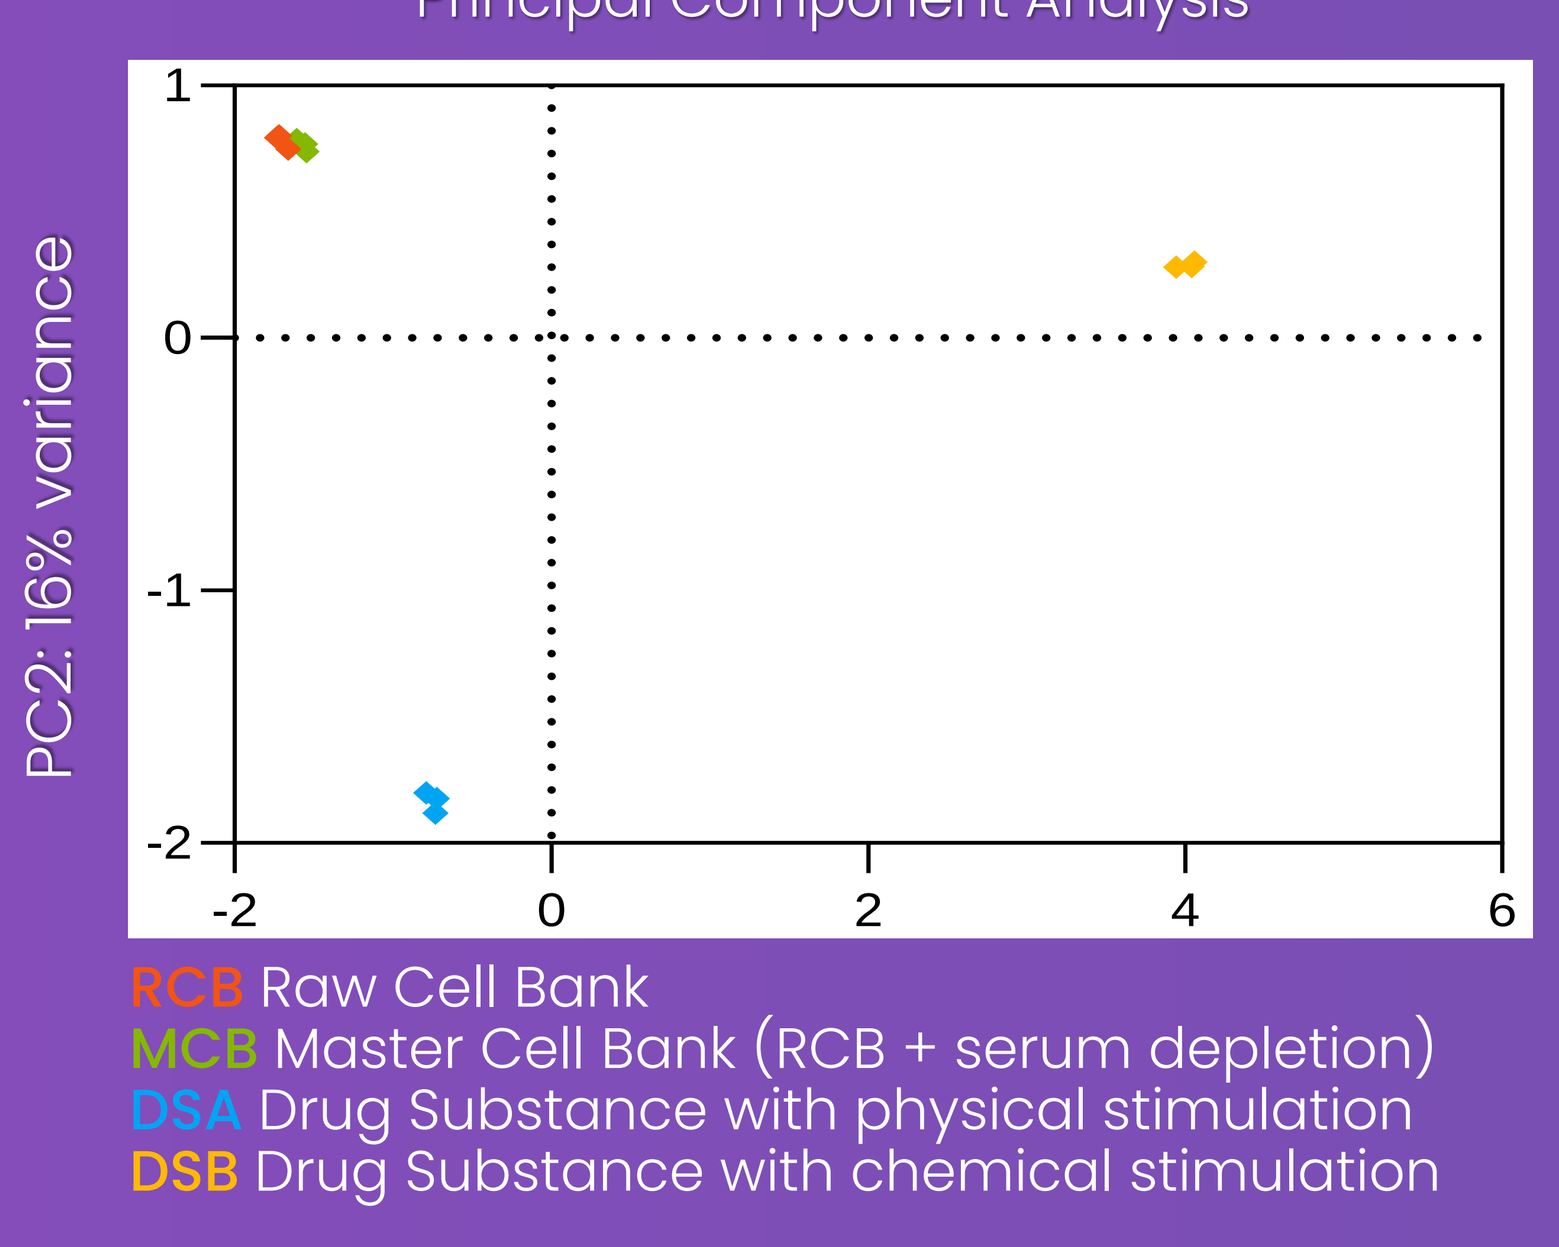
Principal Component Analysis
PC2: 16% variance
RCB Raw Cell Bank
MCB Master Cell Bank (RCB + serum depletion)
DSA Drug Substance with physical stimulation
DSB Drug Substance with chemical stimulation

Fig 2. RNA-seq Analysis

- The transcriptomic and proteomic analysis demonstrate that **each of the physical and chemical stresses modified specific protein expression** which **confirms the rationale for using 3 cell lines with 2 stresses to provide a robust coverage of CRC heterogeneity.**
- **211 Cancer-Related Proteins (CRP)** from the Atlas proteins' database **were identified in 3 batches of STC-1010** including CRC-specific proteins and Tumor-associated antigens linked to tumor plasticity and resistances.

IN-VIVO SYNGENEIC MODEL

- We translated STC Technology to develop a mice surrogate STC-1010 (mSTC-1010)
- Bearing-CRC tumor mice were treated with mSTC-1010

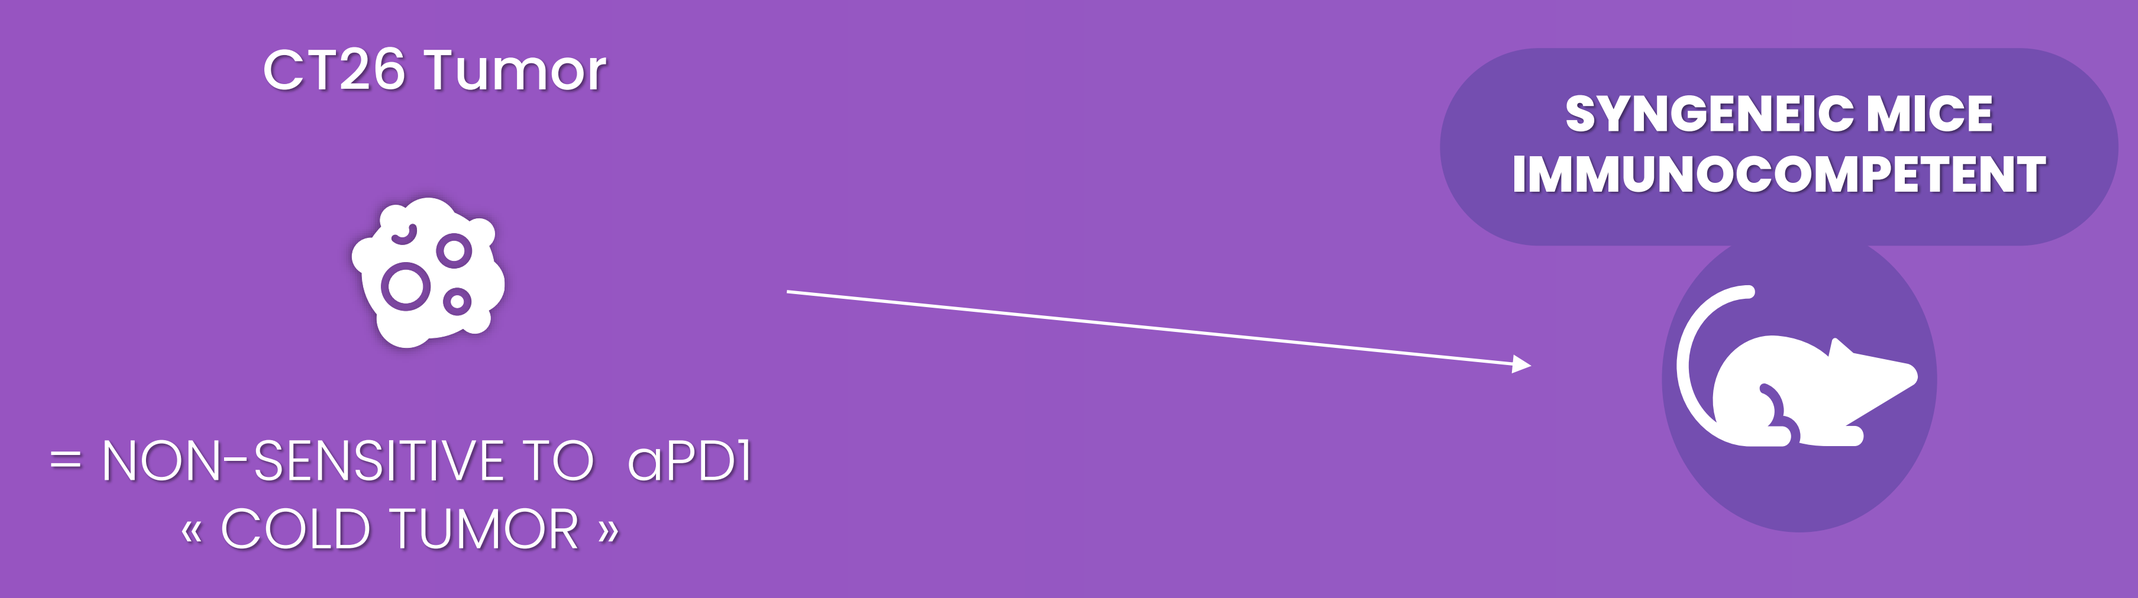
CT26 Tumor
= NON-SENSITIVE TO aPD1
« COLD TUMOR »
SYNGENEIC MICE IMMUNOCOMPETENT

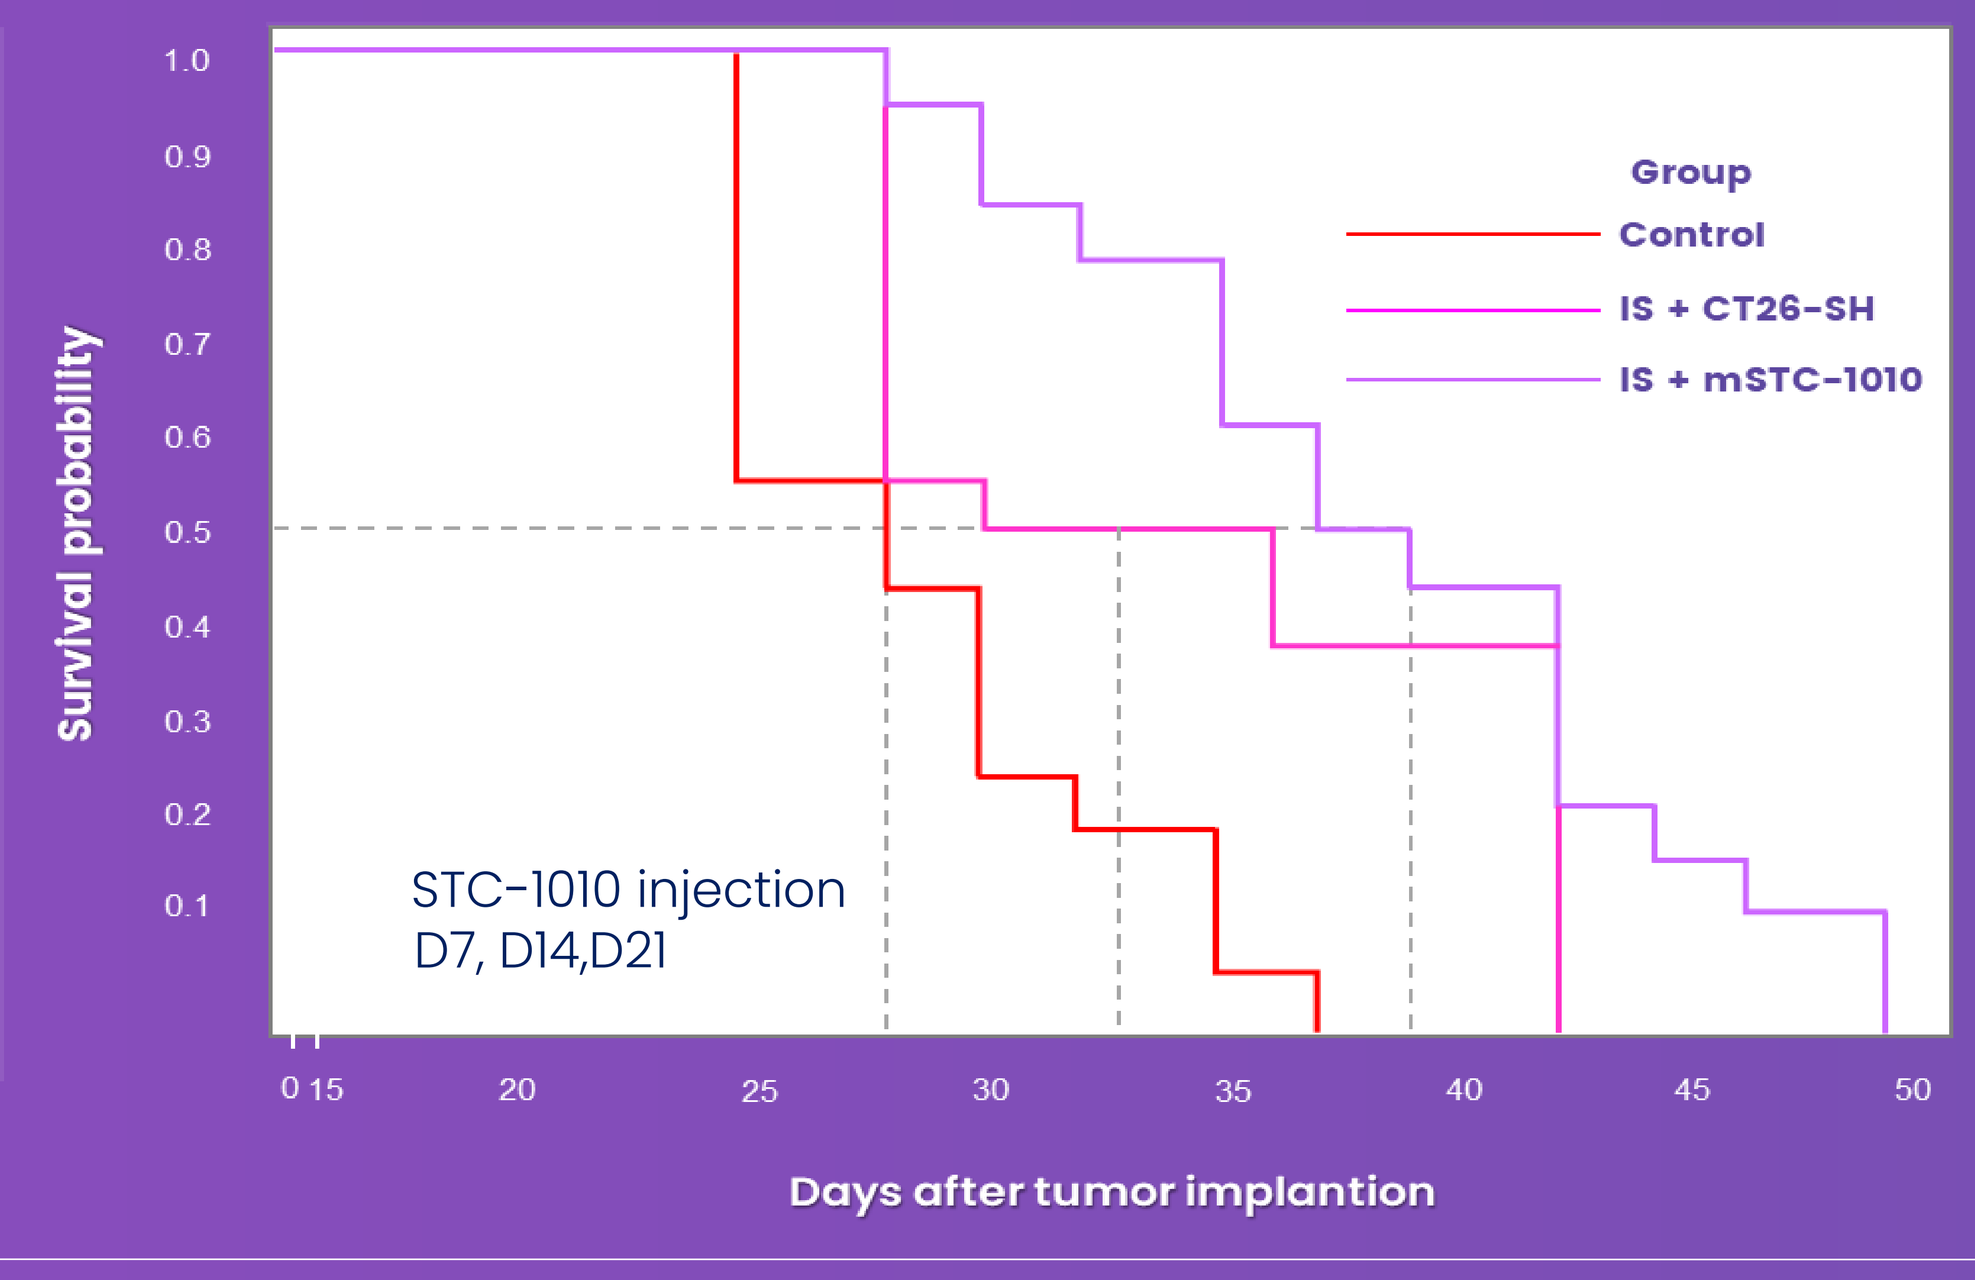
Survival probability
Group
Control
IS + CT26-SH
IS + mSTC-1010
STC-1010 injection
D7, D14,D21
Days after tumor implantion

Fig 3. Kaplan-Meier analysis of the survival of mice CT26 model (n=20 mice / group)

The mSTC-1010 vaccine demonstrates an anti-tumor activity and confirm the hypothesis that 3 cell-line vaccine will be more efficient than 1 ; due to a better representativity of CRC tumor phenotypes

- **Significantly improving the survival of mice bearing tumor, compared to the 1 cell line vaccin and control** (Fig.3)

IS : Immunostimulant cyclophosphamide GM-CFS low dose mSTC-1010: murine STC-1010

IN-OVO CHORIO-ALLANTOÏC MEMBRANE (CAM) MODEL

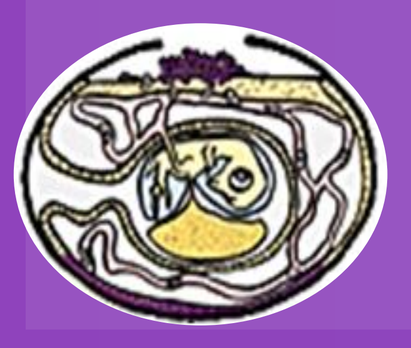

1 Upper Eggshell opening
2 Treatment with STC-1010 At days 2 + 4
3 PBMCs isolation for in ovo treatment
4 PBMCs treatment of eggs bearing HT29 cells at days 2 ,4 & 7
5 Collect and Analysis

STC 1010 induces necrosis of tumor cells and reduces metastatic invasion through the activation of cytotoxic CD8+ Lymphocytes T cells

- **The STC-1010 response is characterized by a significant secretion of IL-12** (p=0.0003), **INFg** (+130,83%, p=0.0185), **and IL-2** (p=0.0033) **reflecting an induction of pro-inflammatory functions of T helper 1 (TH1)** (Fig.5)
- **STC-1010 decreases relative quantity of metastasis vs control** (Fig.6)
- **STC-1010 shows an induction of necrosis score** (Fig.7) **suggesting the implication of IL-12 and INFg secretion to produce massive tumor necrosis.**

Fig.4 STC-1010 impact on cytokine expression vs Ctrl

Fig.5 Metastasis invasion

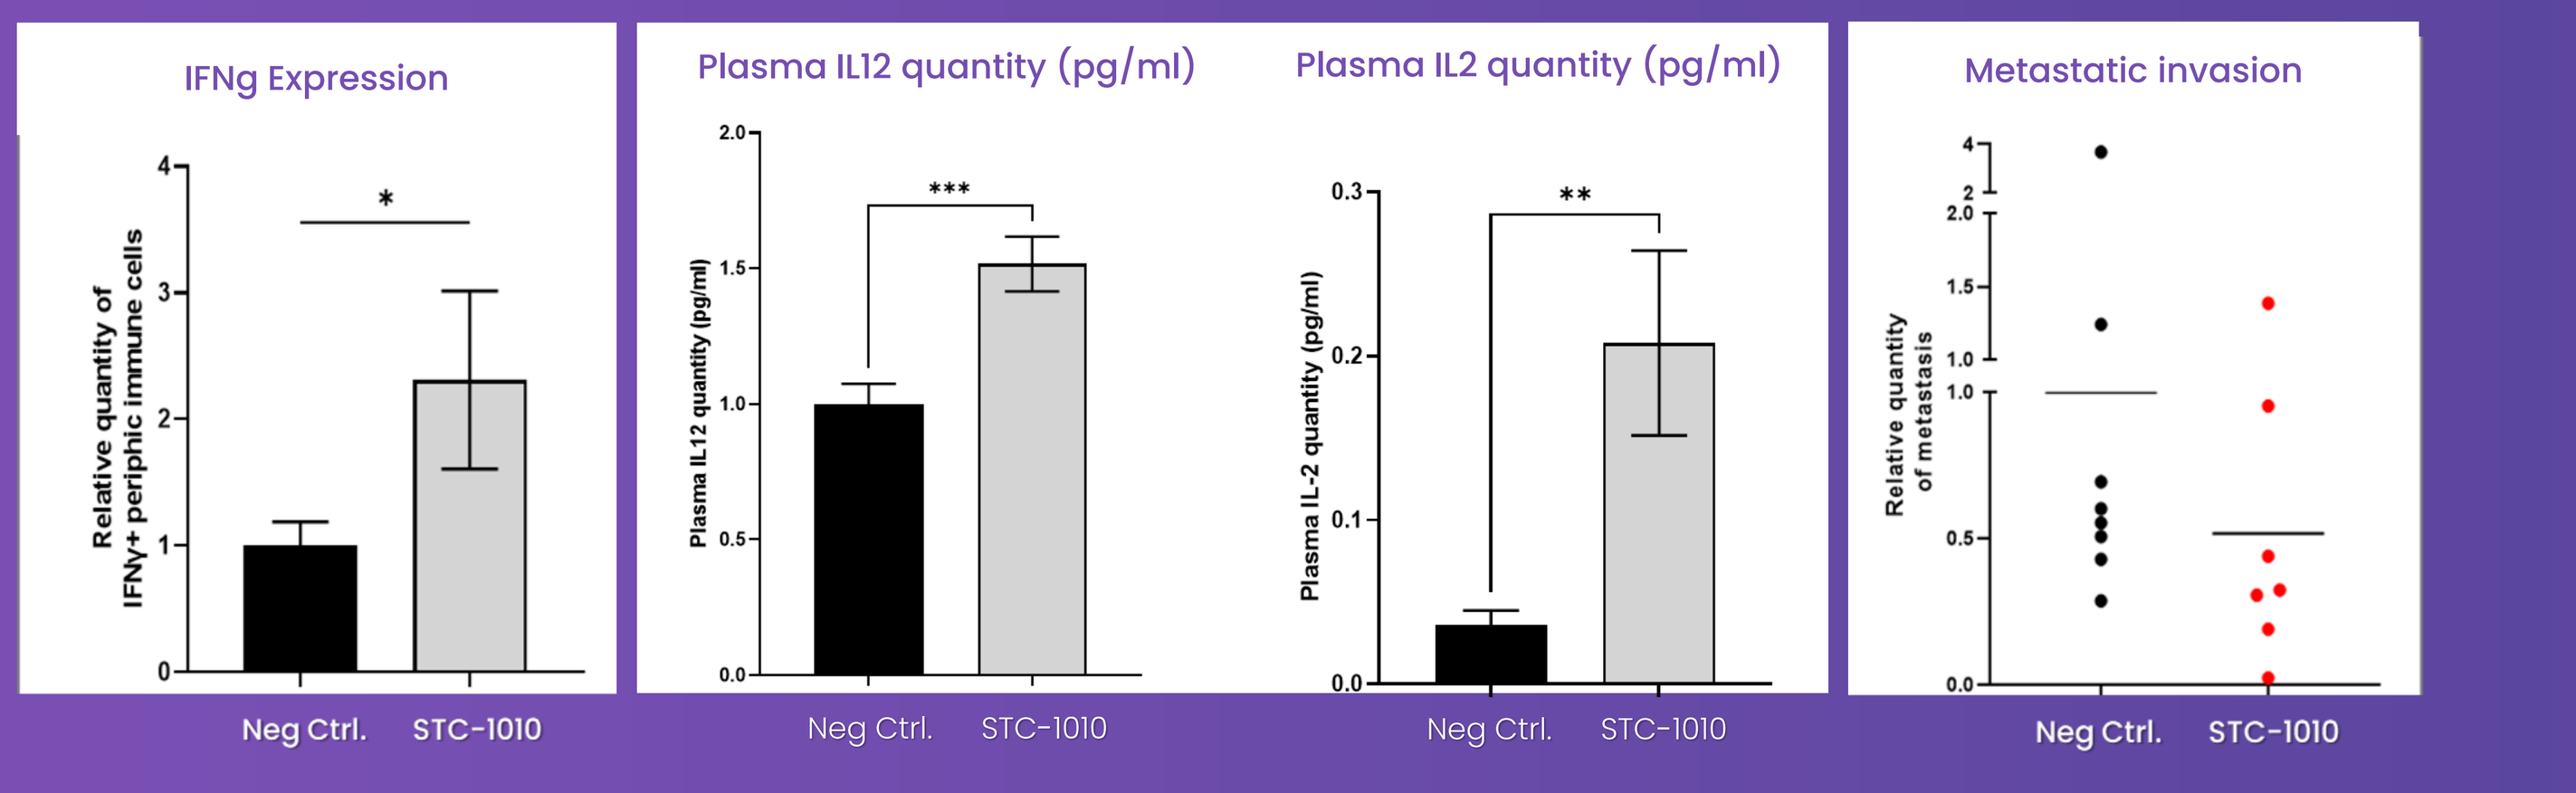
IFNg Expression
Relative quantity of IFNγ+ periphic immune cells
Plasma IL12 quantity (pg/ml)
Plasma IL2 quantity (pg/ml)
Metastatic invasion
Relative quantity of metastasis
Neg Ctrl.
STC-1010

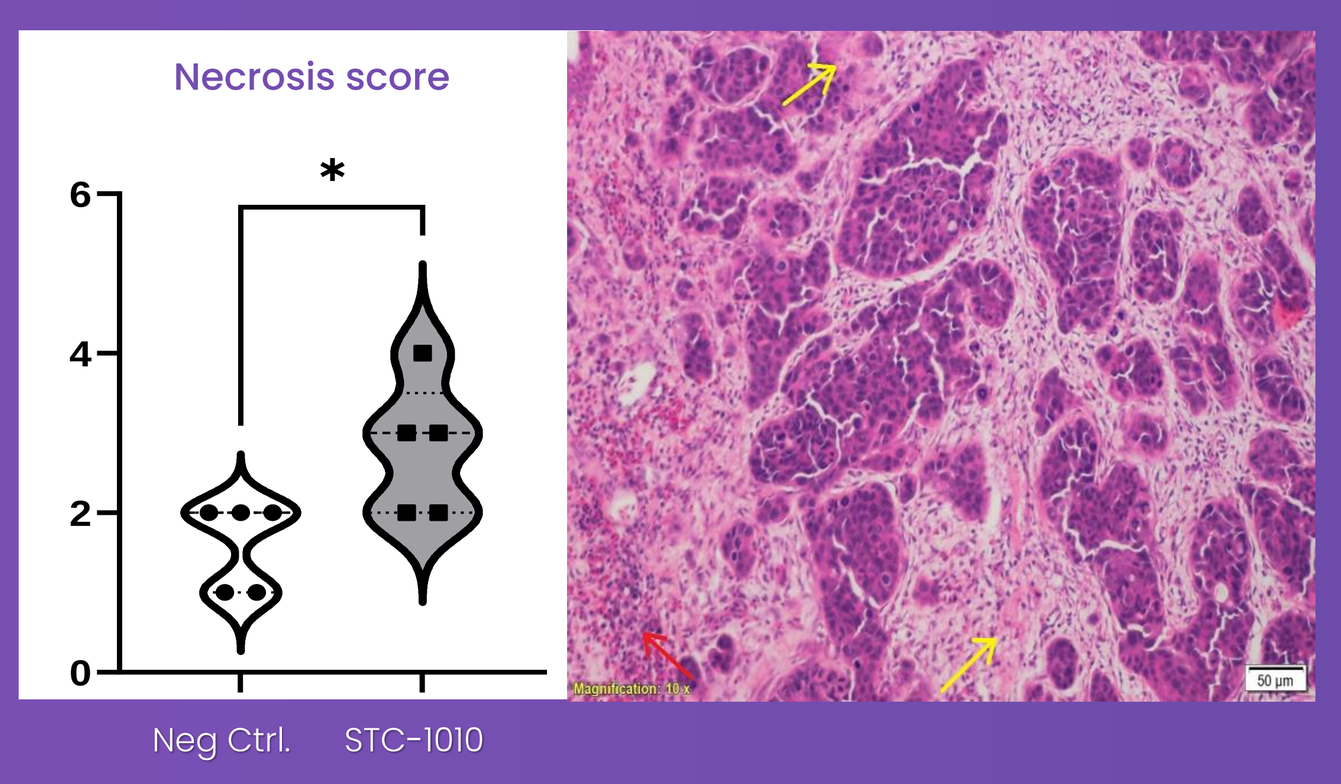
Necrosis score
Neg Ctrl.
STC-1010

Fig.6 Histological necrosis score

Representative photograph:
H&E staining of tumor treated with STC-1010 activated PBMCs (objective magnification x10), Inflammation (red arrow) and necrosis (yellow arrows).

EX-VIVO CO-CULTURE MODEL

1. Activtion of human PBMCs by STC-1010

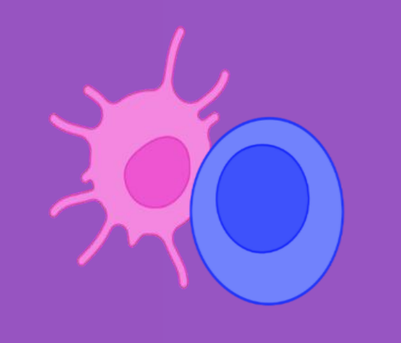

2. Co-Cultrue cell with mature Dendritic Cell and T-Lymphocytes

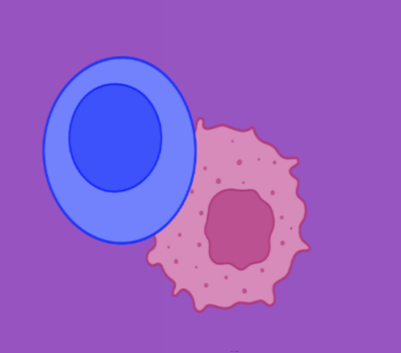

3. Co-culture cell with activated T-CD8 and human CRC cells

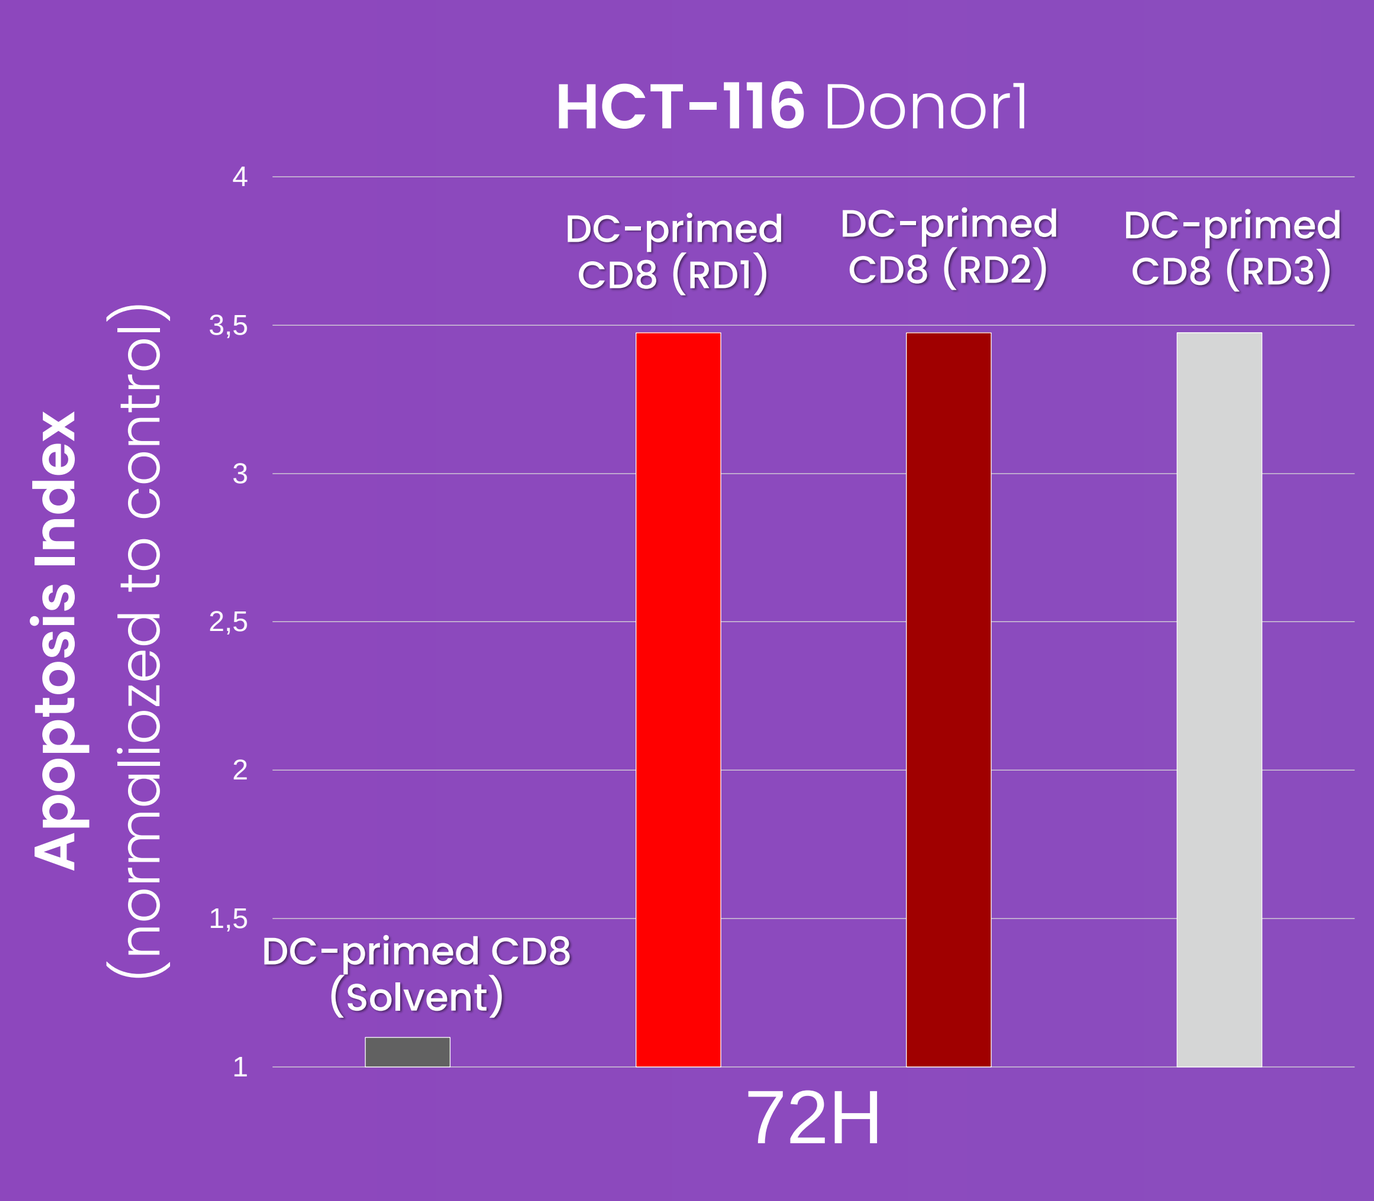
HCT-116 Donor1
Apoptosis Index
(normaliozed to control)
DC-primed CD8 (Solvent)
DC-primed CD8 (RD1)
DC-primed CD8 (RD2)
DC-primed CD8 (RD3)
72H

Fig.7 Apoptosis index of HCT116 tumor cell By STC-1010 (RD1,RD2,RD3) vs ctrl at 72H

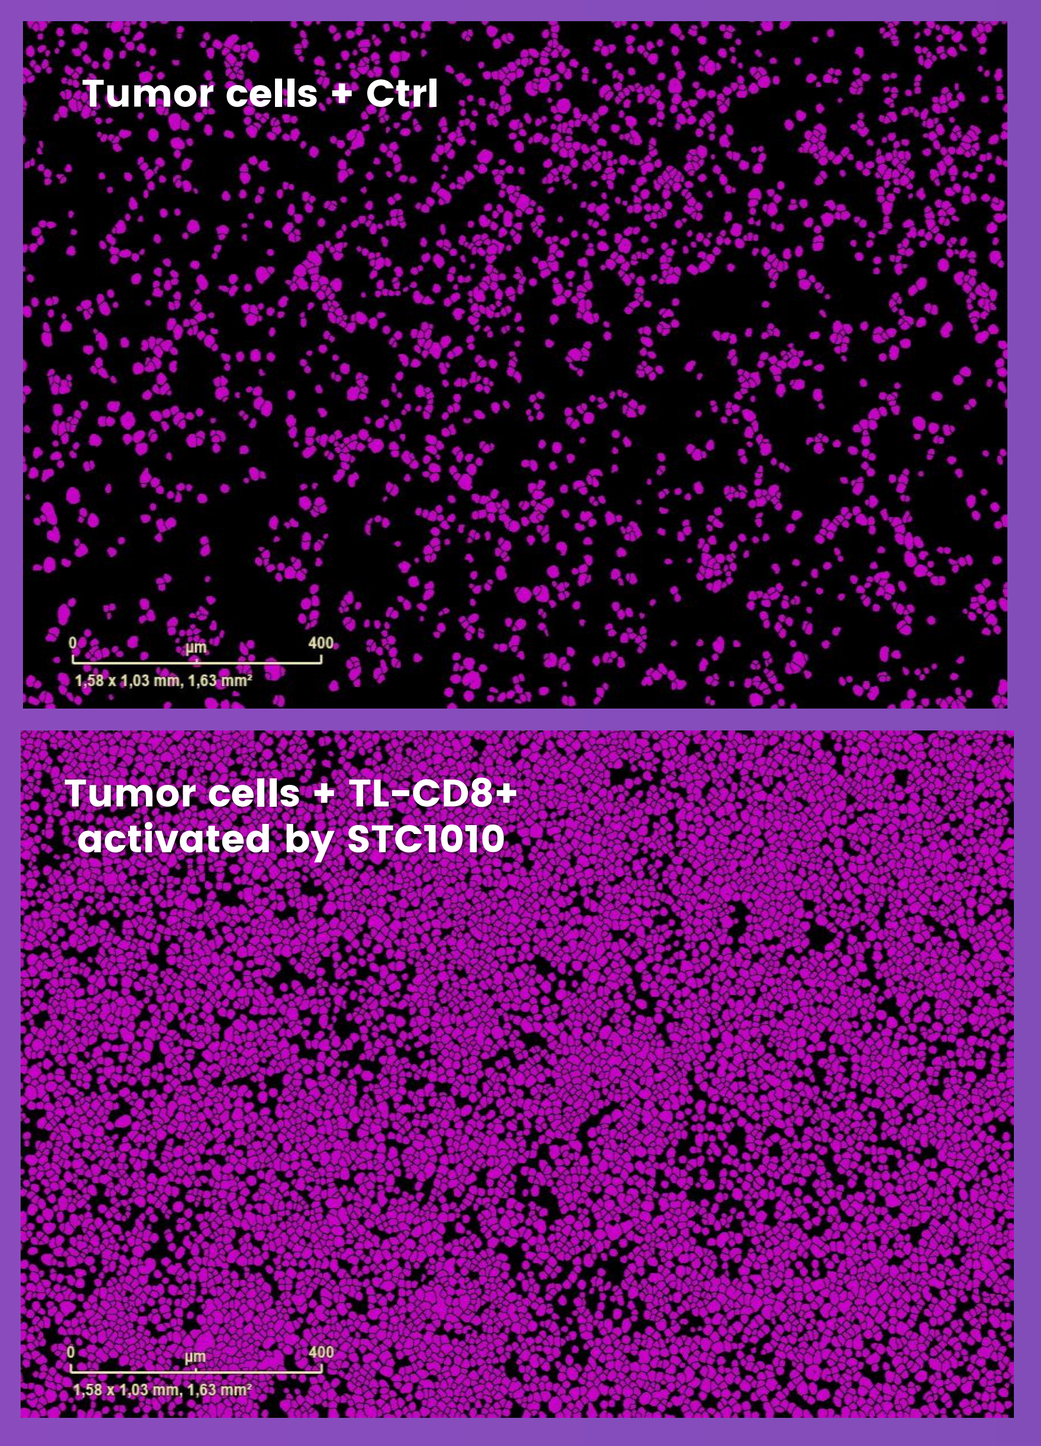
Tumor cells + Ctrl
Tumor cells + TL-CD8+ activated by STC1010

Fig 8 SW620 apoptosis revelation by caspase 3 mediated by DC-primed CD8+ at 72H (purple= apoptotic cells)

STC-1010 induces a robust anti-tumor efficacy via CD8+ apoptotis against CRC tumor

- **STC-1010 increases tumor cell apoptosis with a perfect batch to batch reproductibility** (Fig.7)
- **Strong anti-tumor effect of CD8+T activated by STC-1010 against different CRC cell line with an induction of +62% of SW620 tumor cell apoptosis** (Fig8)

CONCLUSION

Contact : galzeeb@brenus-pharma.com

BASED ON THESE CHARACTERIZATION AND PRE-CLINICAL, IN-OVO AND EX-VIVO EFFICIENCY DATA, STC-1010 TREATMENT WILL SOON BE EVALUATED FOR THE FIRST TIME IN CLINICAL TRIAL IN ADVANCED OR METASTIC STAGE OF COLORECTAL CANCER.